Hybrid DIA and PRM Acquisition Enables In-Depth Host Cell Protein Profiling with Relative Quantitation of Problematic Targets

Christopher Sauer[1], Abby Chiang[2], Elsa Gorre[1], Jae Choi[3], Nikki Jarrett[3], Bhavin Patel[3], Min Du[4], Andrew Mahan[1], Hirsh Nanda[1]

[1]Johnson & Johnson Innovative Medicine, Spring House, PA;
[2]Protein Metrics, LLC, Boston, MA;
[3]Thermo Fisher Scientific, Rockford, IL;
[4]Thermo Fisher Scientific, San Jose, CA

Introduction

- The identification and quantitative analysis of host cell proteins is made difficult by the large dynamic range of analytes, especially compared to the biotherapeutic.
- Antibody Depletion is evaluated here along with Native Digestion to improve sensitivity and depth of coverage.
- To further interrogate a targeted panel of problematic HCPs, without sacrificing profiling abilities, a Hybrid DIA/PRM workflow is used

Cell Engineering and Analytical Sciences

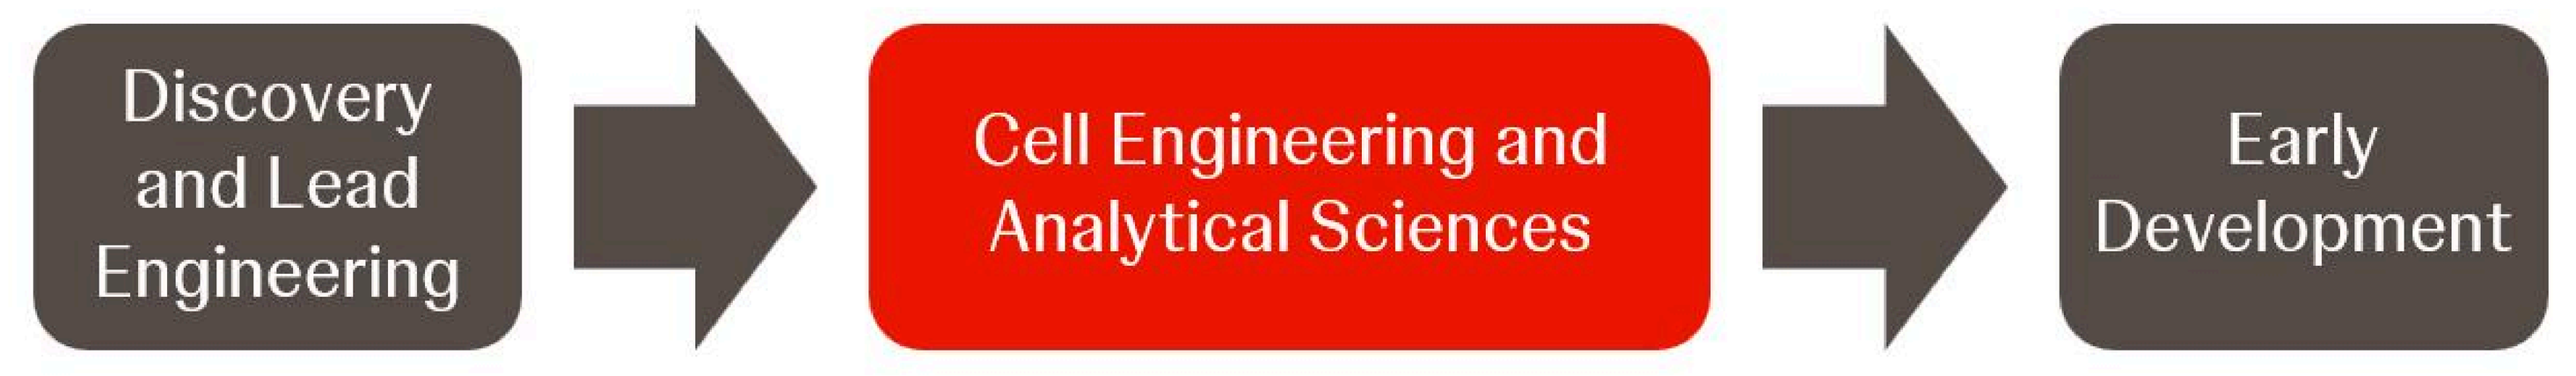
Discovery and Lead Engineering
Cell Engineering and Analytical Sciences
Early Development

Cell Engineering and Analytical Sciences (CEAS) sits at the entry point for biotherapeutics entering the manufacturing pipeline from lead engineering.

- Diverse portfolio of mAbs, multi-specifics, and ADCs

Methods

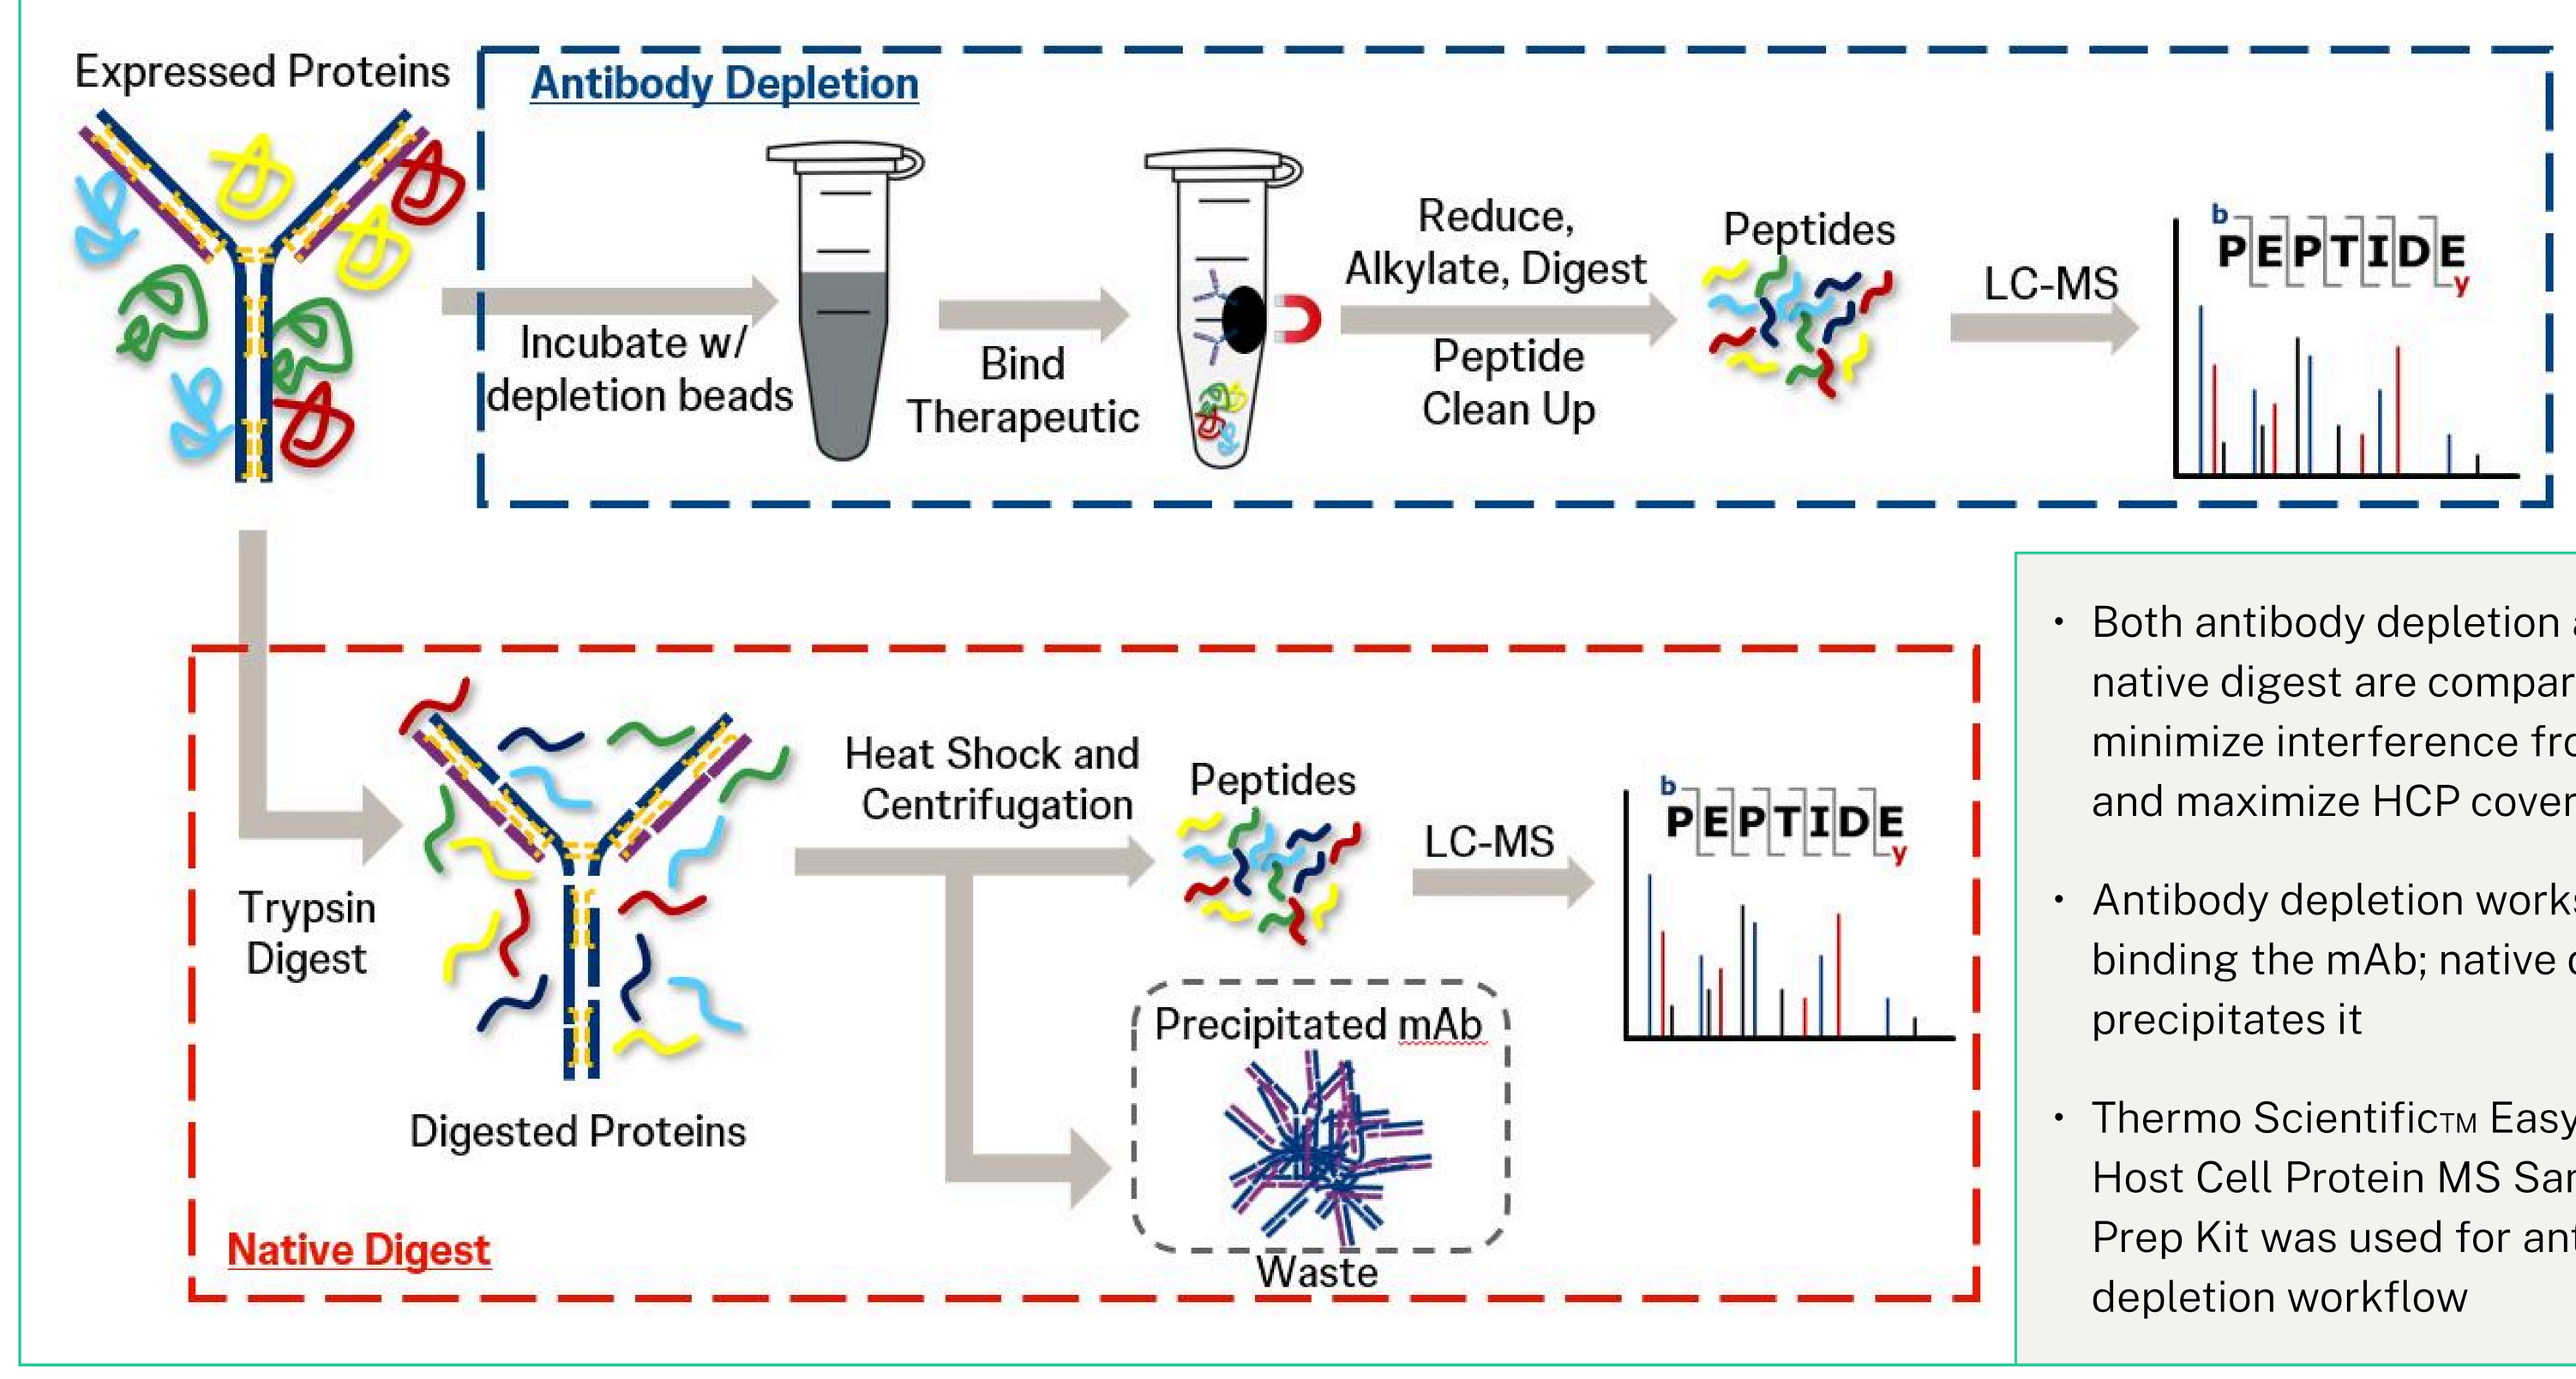
Expressed Proteins
Antibody Depletion
Incubate w/ depletion beads
Bind Therapeutic
Reduce, Alkylate, Digest
Peptide Clean Up
Peptides
LC-MS
PEPTIDE
Trypsin Digest
Digested Proteins
Heat Shock and Centrifugation
Peptides
LC-MS
PEPTIDE
Precipitated mAb
Waste
Native Digest

- Both antibody depletion and native digest are compared to minimize interference from mAb and maximize HCP coverage[1]
- Antibody depletion works by binding the mAb; native digest precipitates it
- Thermo Scientific™ EasyPep™ Host Cell Protein MS Sample Prep Kit was used for antibody depletion workflow

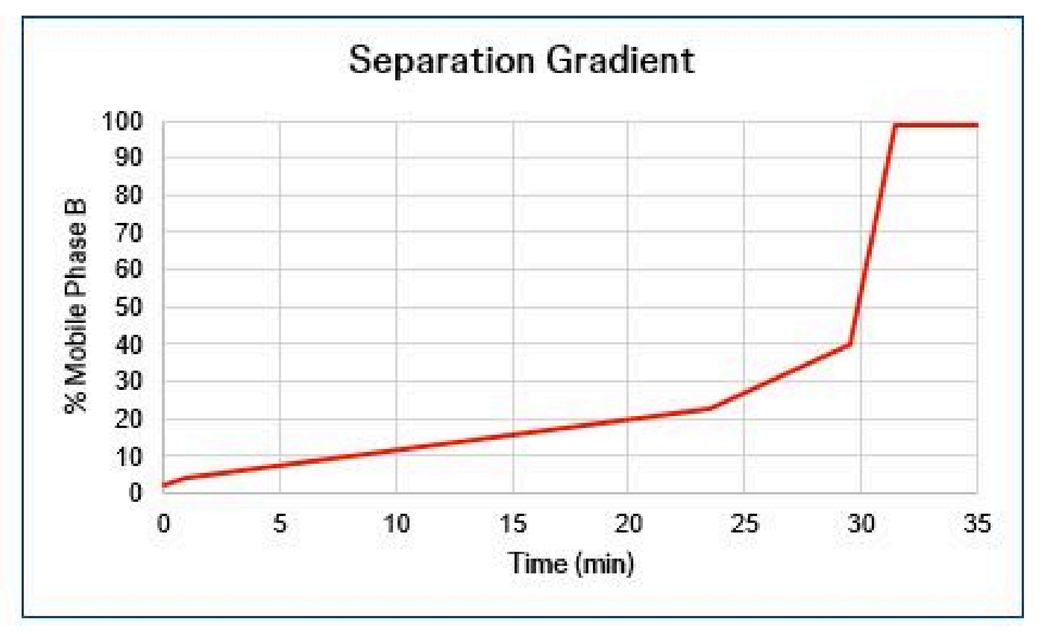
Separation Gradient
% Mobile Phase B
Time (min)

Mobile Phase A: 0.1% FA in H_2O

Mobile Phase B: 0.1% FA in ACN

Flow Rate: 0.3 mL/min

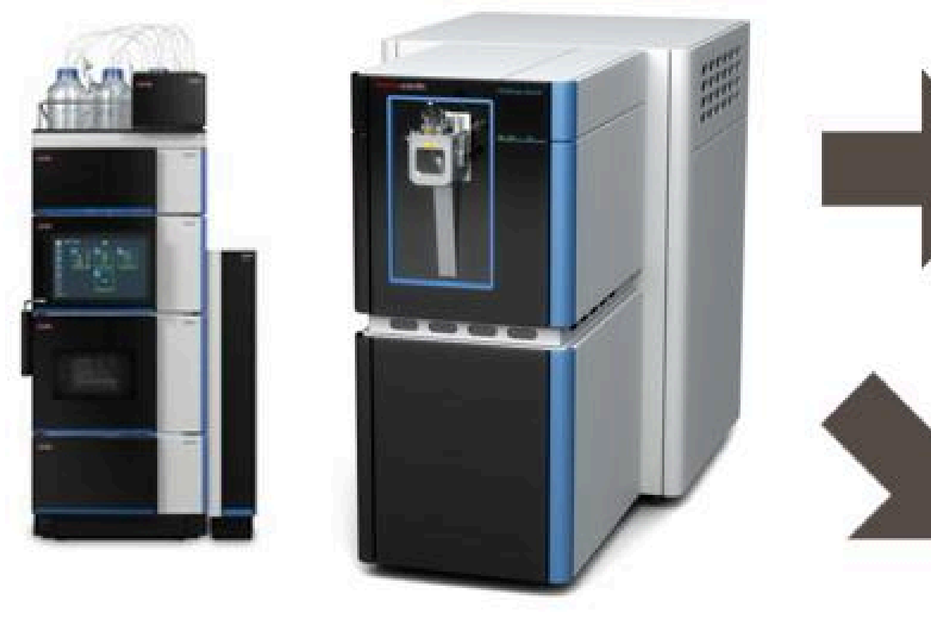

Analytical flow RPLC coupled to Thermo Orbitrap Astral Mass Spectrometer[2]

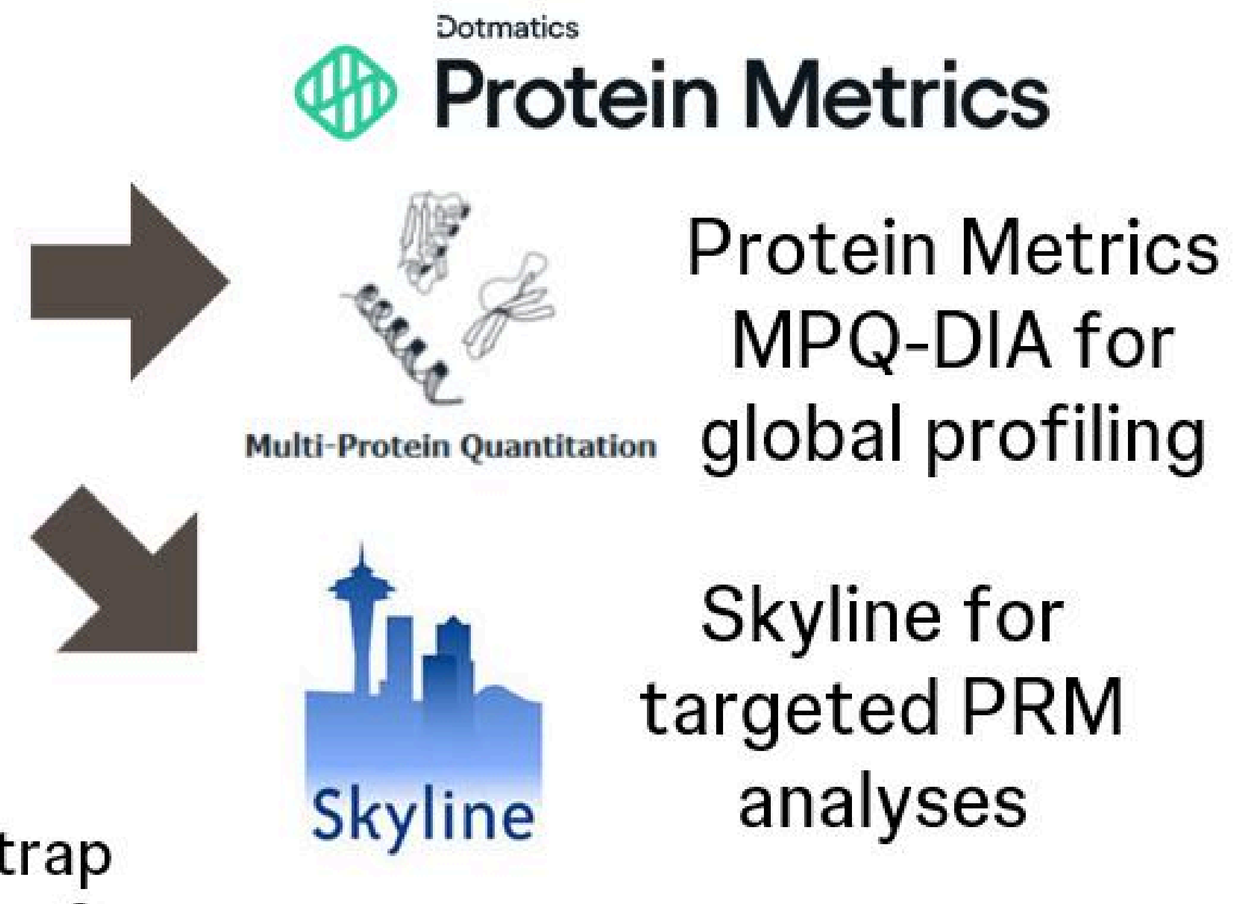
Dotmatics
Protein Metrics
Multi-Protein Quantitation
Protein Metrics MPQ-DIA for global profiling
Skyline
Skyline for targeted PRM analyses

Workflow Optimization

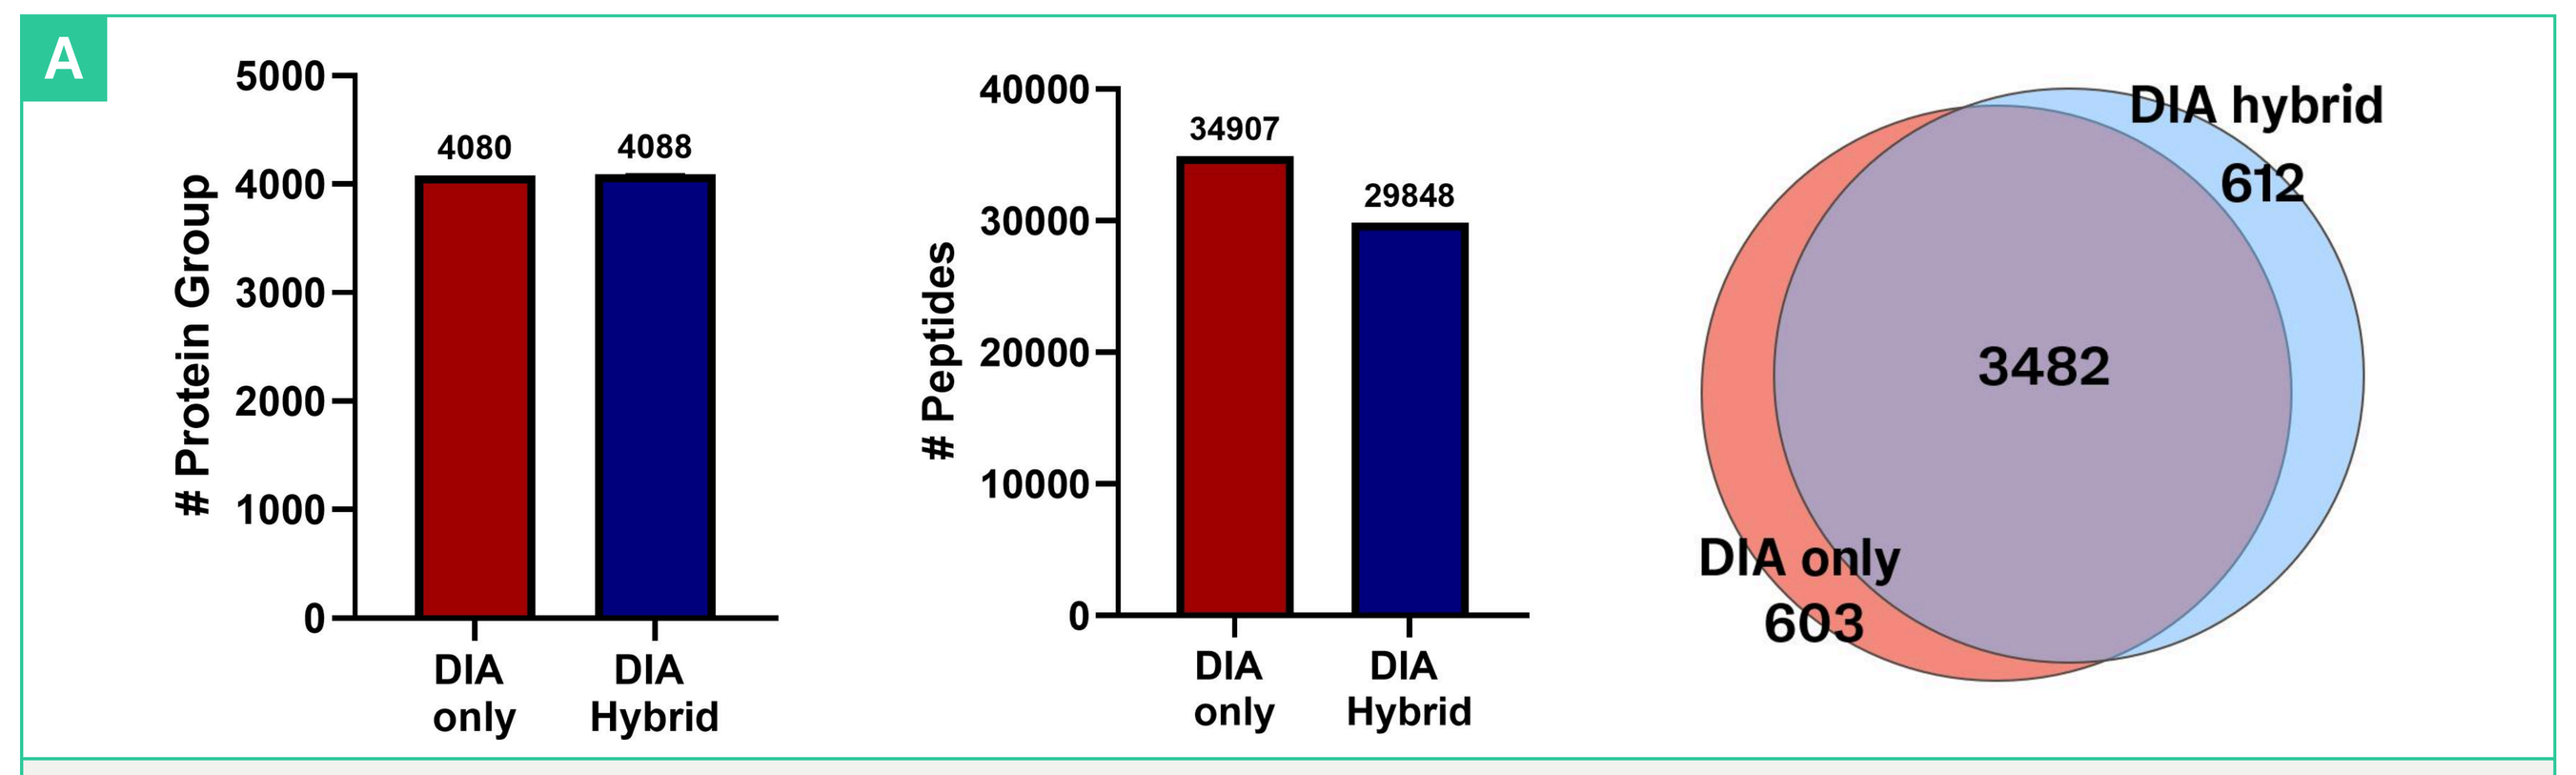
A
Protein Group
4080
4088
DIA only
DIA Hybrid
Peptides
34907
29848
DIA only
DIA Hybrid
DIA hybrid
612
3482
DIA only
603

- DIA vs. hybrid DIA/PRM on an Orbitrap Astral MS with 200ng Hela digest (n=3).
- The DIA-only method identified a higher total number of peptides, but there was no significant difference in total protein identifications between methods.

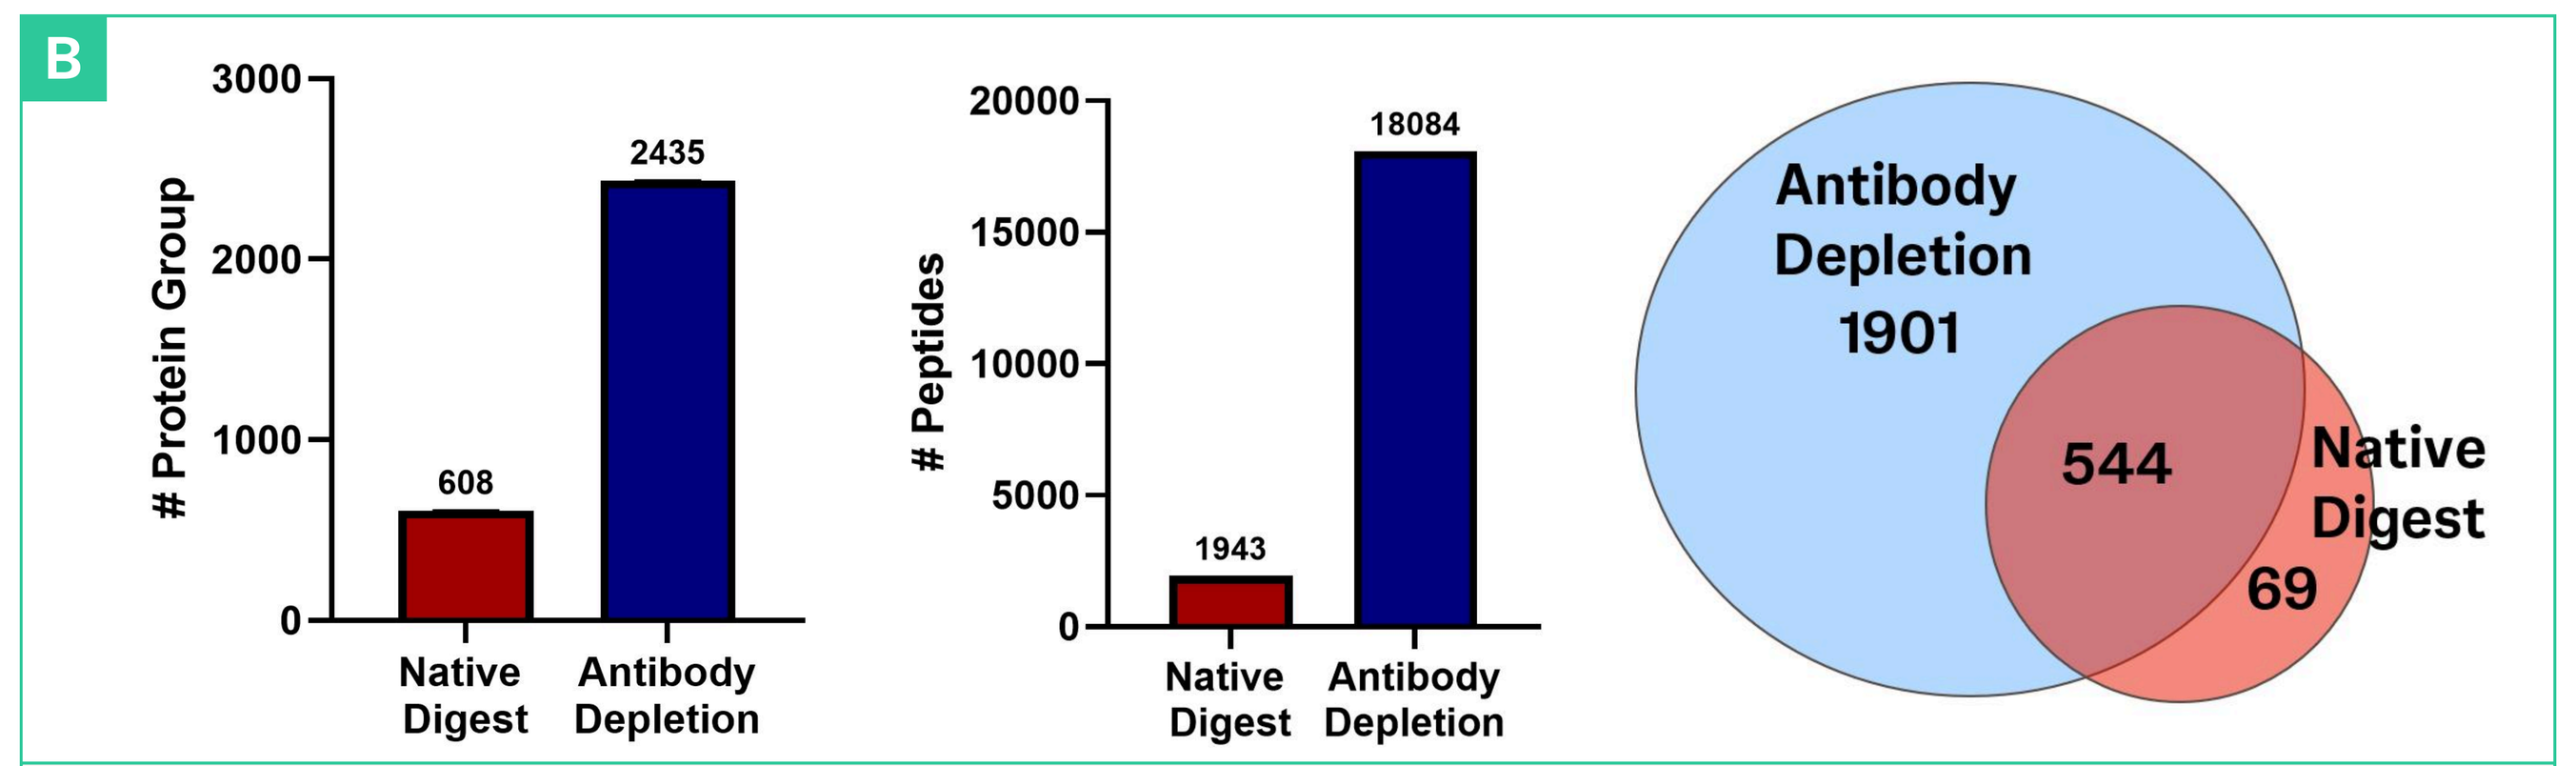
B
Protein Group
608
2435
Native Digest
Antibody Depletion
Peptides
1943
18084
Native Digest
Antibody Depletion
Antibody Depletion
1901
544
Native Digest
69

- Clear Harvest samples (purification stage 0; n = 3) were processed by either native digestion or antibody depletion workflow and analyzed using a hybrid DIA/PRM acquisition.
- The antibody depletion workflow yielded a significantly higher number of identified proteins and peptides compared with the native digest.

HCP Global Profiling via DIA MS

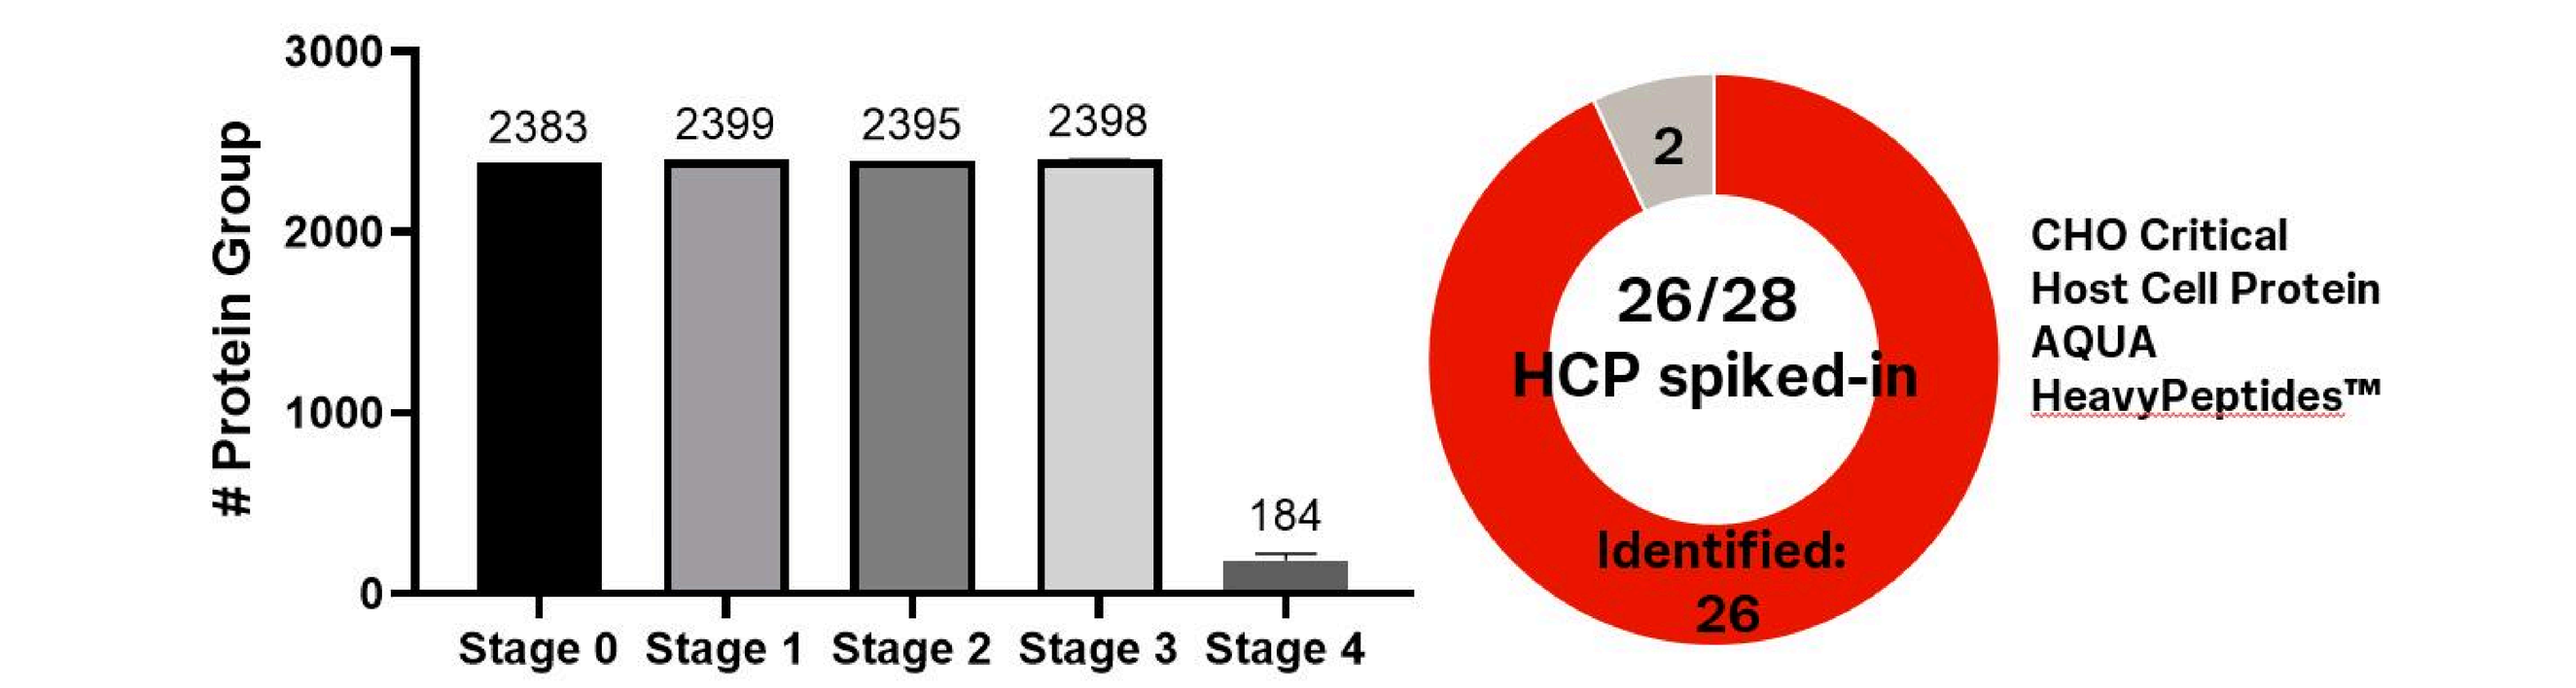
Protein Group
2383
2399
2395
2398
184
Stage 0
Stage 1
Stage 2
Stage 3
Stage 4
2
26/28
HCP spiked-in
Identified:
26
CHO Critical Host Cell Protein AQUA HeavyPeptides™

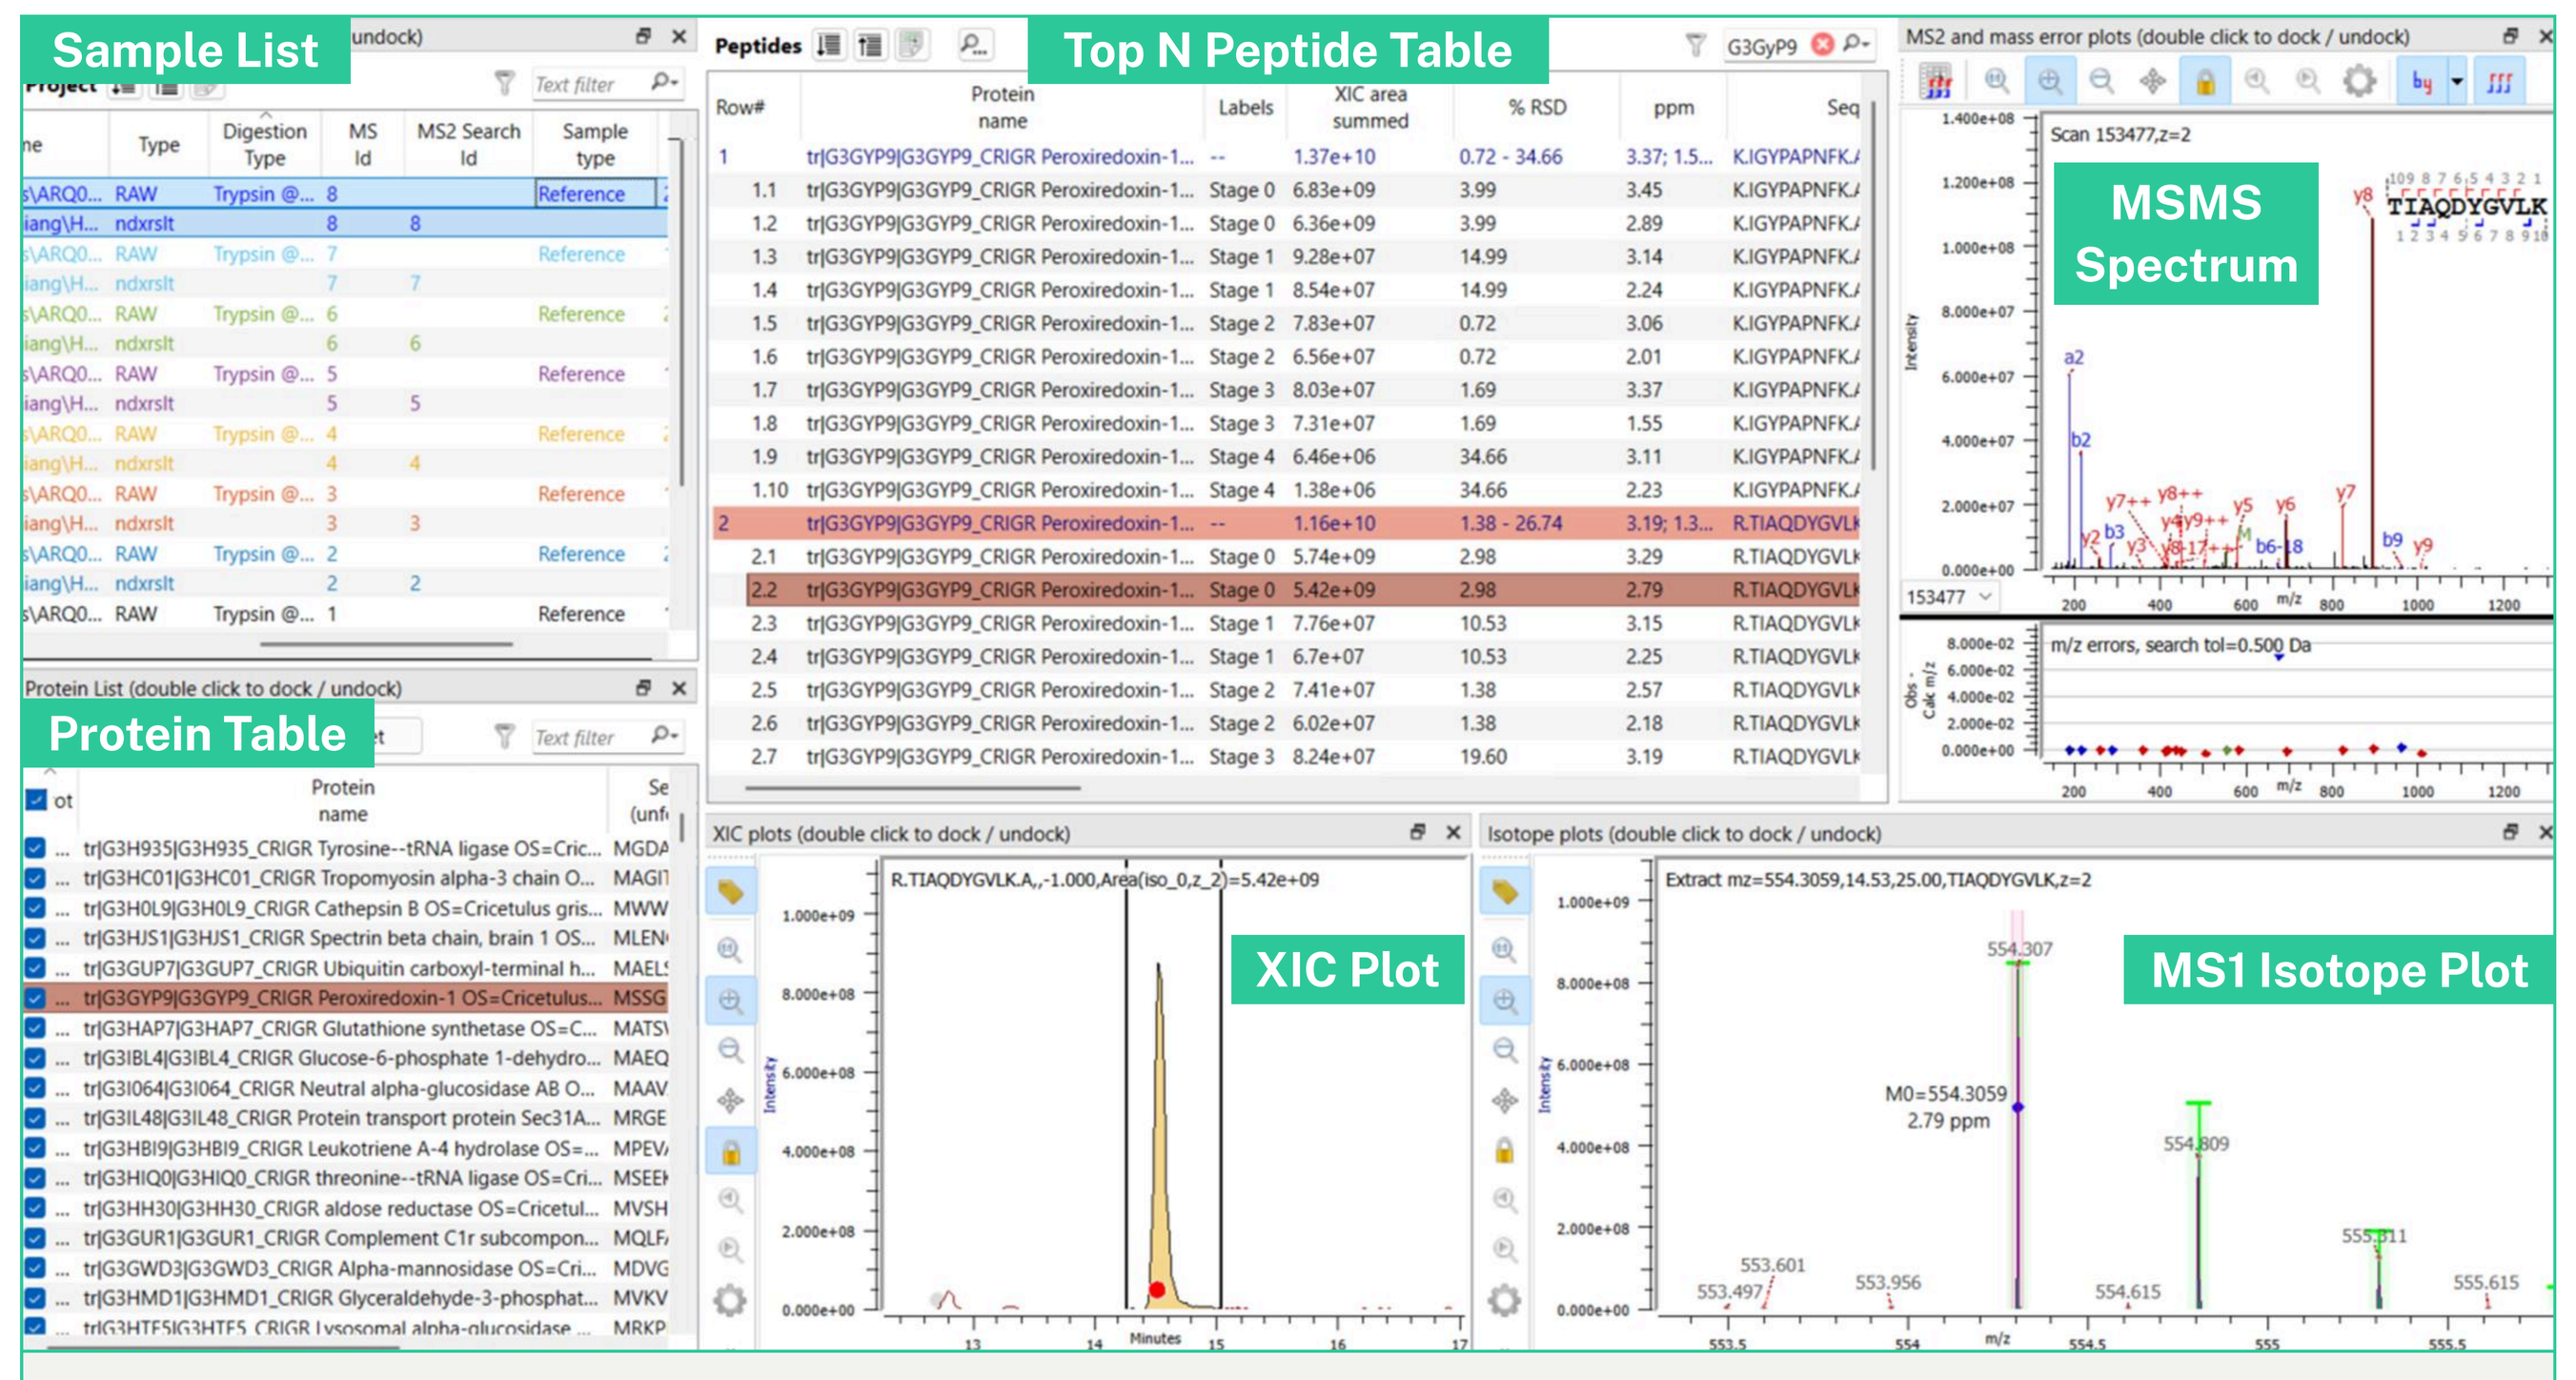
Sample List
Top N Peptide Table
MSMS Spectrum
Protein Table
XIC Plot
MS1 Isotope Plot

- The total number of HCPs detected at each purification stage is shown in the top panel.
- Samples were processed using the antibody depletion workflow and analyzed with a hybrid DIA/PRM acquisition.
- All 26 spiked-in target HCPs were detected in the global profiling dataset and showed trends consistent with the targeted analysis.

Targeted Analysis of Problematic HCPs

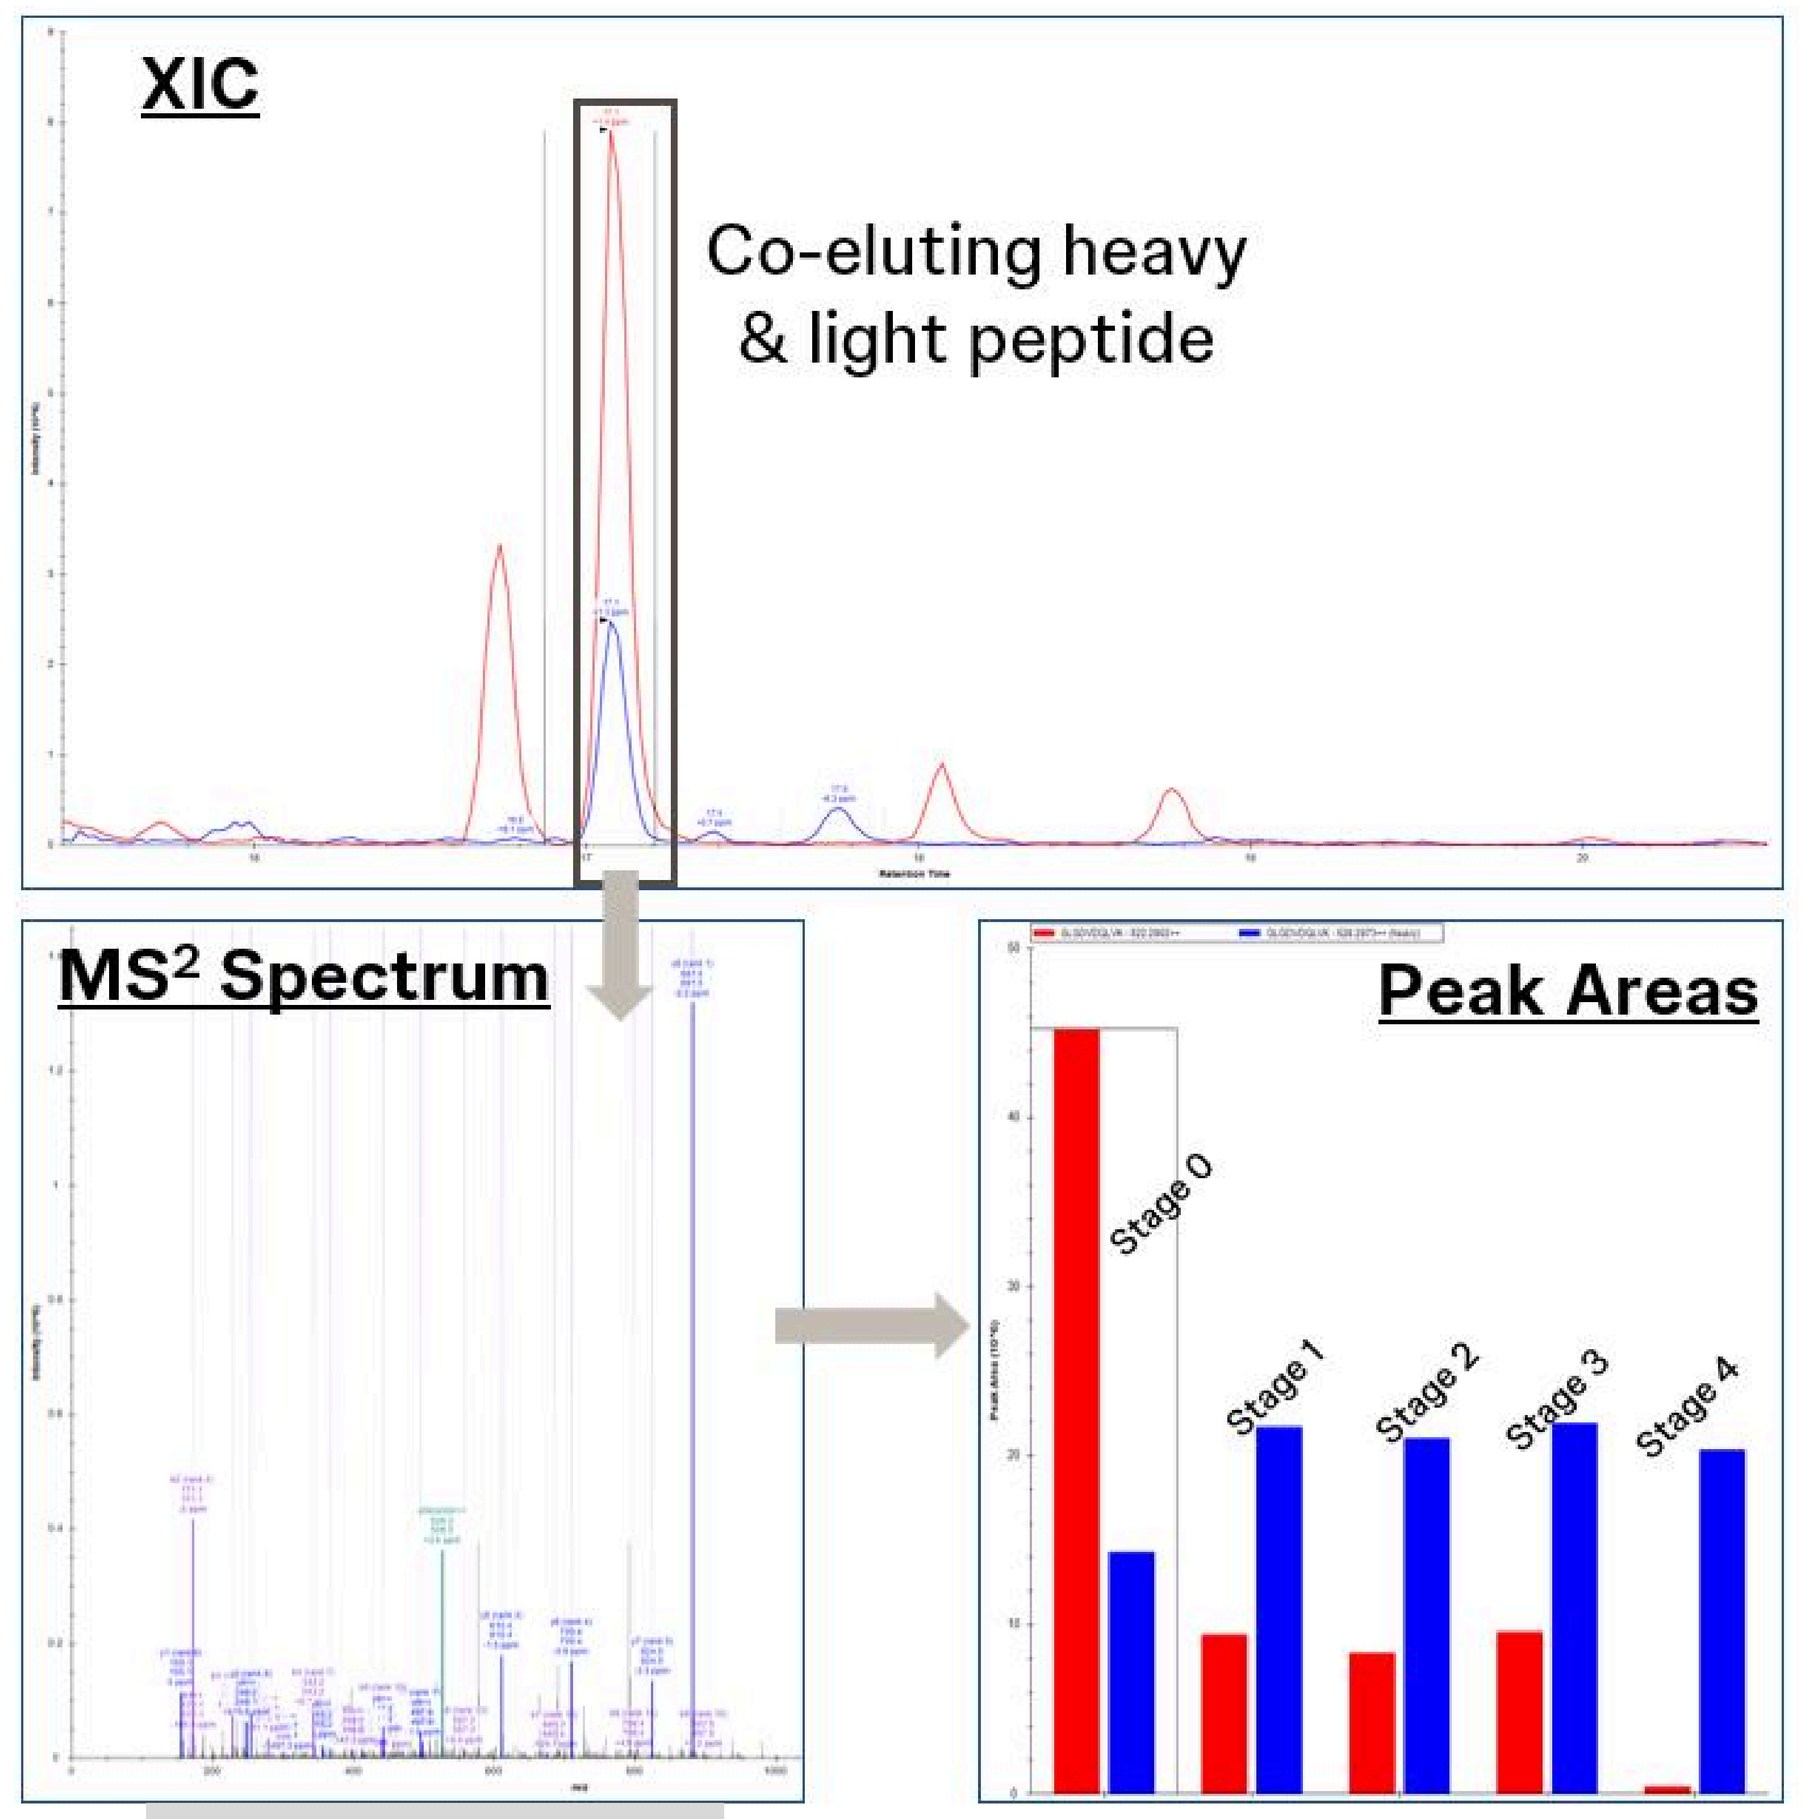
XIC
Co-eluting heavy & light peptide
MS2 Spectrum
Peak Areas
Stage 0
Stage 1
Stage 2
Stage 3
Stage 4

- Targeted panel of HCPs identified and quantified based on spiked in heavy peptides[3]
- High sequence coverage and retention time alignment allow for low level quantitation and tracking HCPs across purification stages

Targeted HCP Panel

78 kDa glucose regulated protein(GRP78, BiP)	Lipoprotein Lipase (LPL)
Alpha-enolase (2-phospho-D-glycerate hydro-lyase)	Lysosomal Acid Lipase (LAL)
Annexin A5 (ANXA5)	Lysosomal Phospholipase A2 (LPLA2)
C-C motif chemokine (CCL)	Matrix metalloproteinase-19 (MMP-19)
C-X-C motif chemokine 3 (CXCL3)	Peptidyl-prolyl cis-trans isomerase A (PPIA)
Carboxylesterase 1-like protein, Liver (CES1)	Peroxiredoxin-1 (PRDX1)
Liver carboxylesterase 4	Phospholipase B-like 2 (PLBL2)
Carboxypeptidase D (Cpd)	Procollagen-lysine 2-oxoglutarate 5-deoxygenase_1 (PLOD1)
Cathepsin B (CatB)	Protein disulfide isomerase (PDI)
Cathepsin D (CatD)	Protein S100-A6 (S10A6)
Cathepsin L (CatL)	Pyruvate Kinase (PK)
Cathepsin Z (CatZ)	Serine protease (HTRA1)
Clusterin (CLU)	Sialate o-acetylesterase (SIAE)
Glutathione S-transferase P 1 (GSTP1)	Transforming Growth Factor-b1 (TGF-β1)

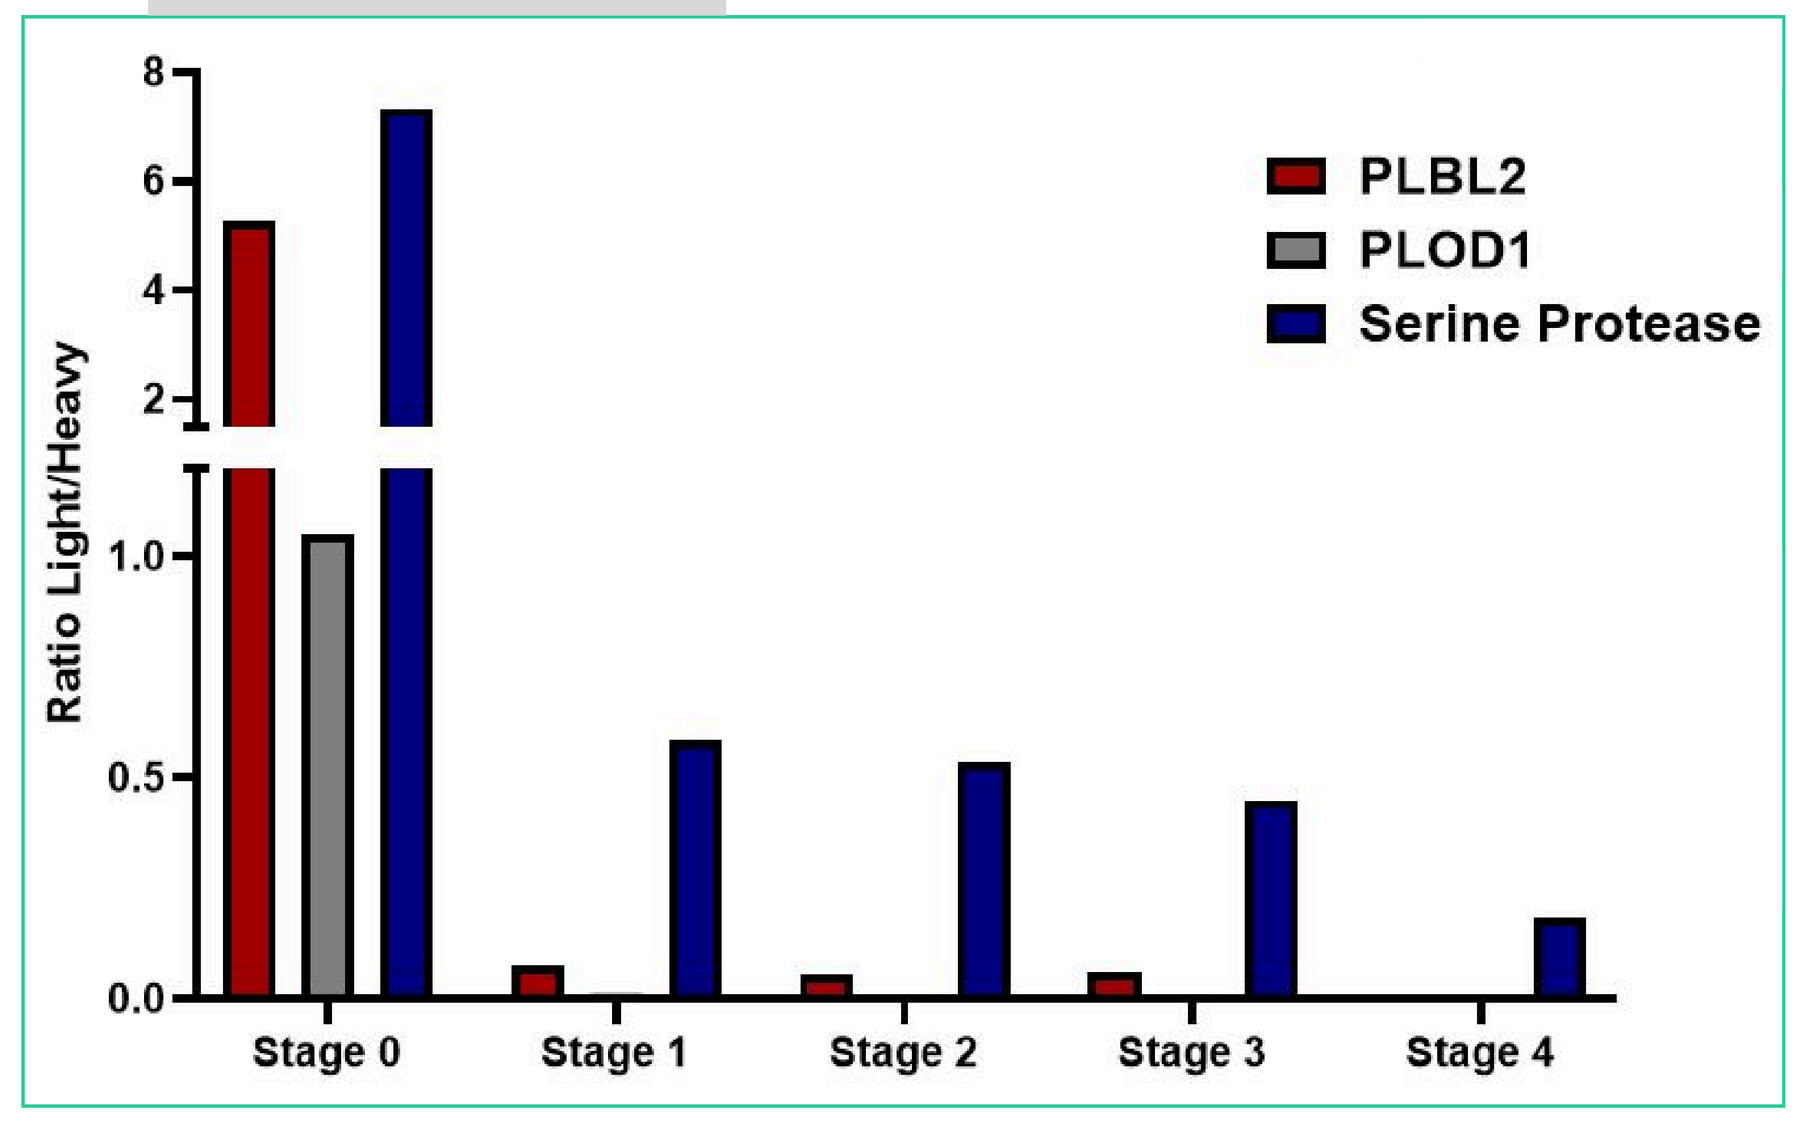
Ratio Light/Heavy
PLBL2
PLOD1
Serine Protease
Stage 0
Stage 1
Stage 2
Stage 3
Stage 4

- Example data for selected HCPs: Peak areas relative to the heavy labeled peptide show high rates of clearance at Stage 1.
- By stage 4, many HCPs are approaching LODs
- This trend is common for most HCPs

Conclusions

- ✓ Hybrid DIA/PRM achieved comparable proteome coverage versus DIA-only in a complex sample, while enabling targeted PRM confirmation of selected analytes.
- ✓ Antibody depletion resulted in greater depth of coverage for the HCP profiles compared to native digestion
- ✓ Heavy labeled peptides from known high risk HCPs enabled retention time alignment, MS2 validation, and relative quantitation of the wild type proteins. This can be applied to many studies, such as evaluating clearance of HCPs at different purification stages.

References

1. Huang L, Wang N, Mitchell CE, Brownlee TJ, Maple SR, De MR. A Novel Sample Preparation for Shotgun Proteomics Characterization of HCPs in Antibodies. 2017;89(10):5436-5444. doi:https://doi.org/10.1021/acs.analchem.7b00304
2. Kiyonami R, Melani R, Chen Y, Leon AD, Du M. Applying UHPLC-HRAM MS/MS Method to Assess Host Cell Protein Clearance during the Purification Process Development of Therapeutic mAbs. International Journal of Molecular Sciences. 2024; 25(17):9687. https://doi.org/10.3390/ijms25179687
3. Jones, M. , Palackal, N. , Wang, F. , Gaza-Bulseco, G. , Hurkmans, K. , Zhao, Y. , Chitikila, C. , Clavier, S. , Liu, S. , Menesale, E. , Schonenbach, N. S. , Sharma, S. , Valax, P. , Waerner, T. , Zhang, L. , & Connolly, T. (2021). “High-risk” host cell proteins (HCPs): A multicompany collaborative view. Biotechnology Bioengineering. 118, 2870–2885. https://doi.org/10.1002/bit.27808

The authors declare no competing financial interest.

Johnson&Johnson Protein Metrics Thermo Fisher SCIENTIFIC Therapeutics Development & Supply